CAYMAN BIOANALYSIS SERVICES

Lipidomics in Cells, Plasma, and Tissues

Bioactive Lipid Profiling

Lipids are involved in an astonishing variety of physiological and pathological processes and are the object of intense research efforts from scientists around the world. However, the analytical tools and know-how to perform lipidomics analyses are not readily available to many researchers.

With Cayman Contract Services, your lipidomics study is in the hands of experts who have many decades of collective experience in lipid synthesis, purification, and characterization.

Cayman's state-of-the art mass spectrometry laboratories are equipped with triple-quadrupole and accurate-mass Orbitrap LC-MS systems. Enabling us to offer bioanalysis services customized for your specific needs.

Our analysis service offers the sensitivity, efficiency, and specificity needed to identify and quantify these biologically relevant lipids:

- Ceramides
- Cholesterol Esters
- Eicosanoids
- Glycerolipids
- Oxylipins
- Phospholipids
- Short-Chain Fatty Acids
- Sphingolipids
- Urinary Metabolites

Standard Workflow

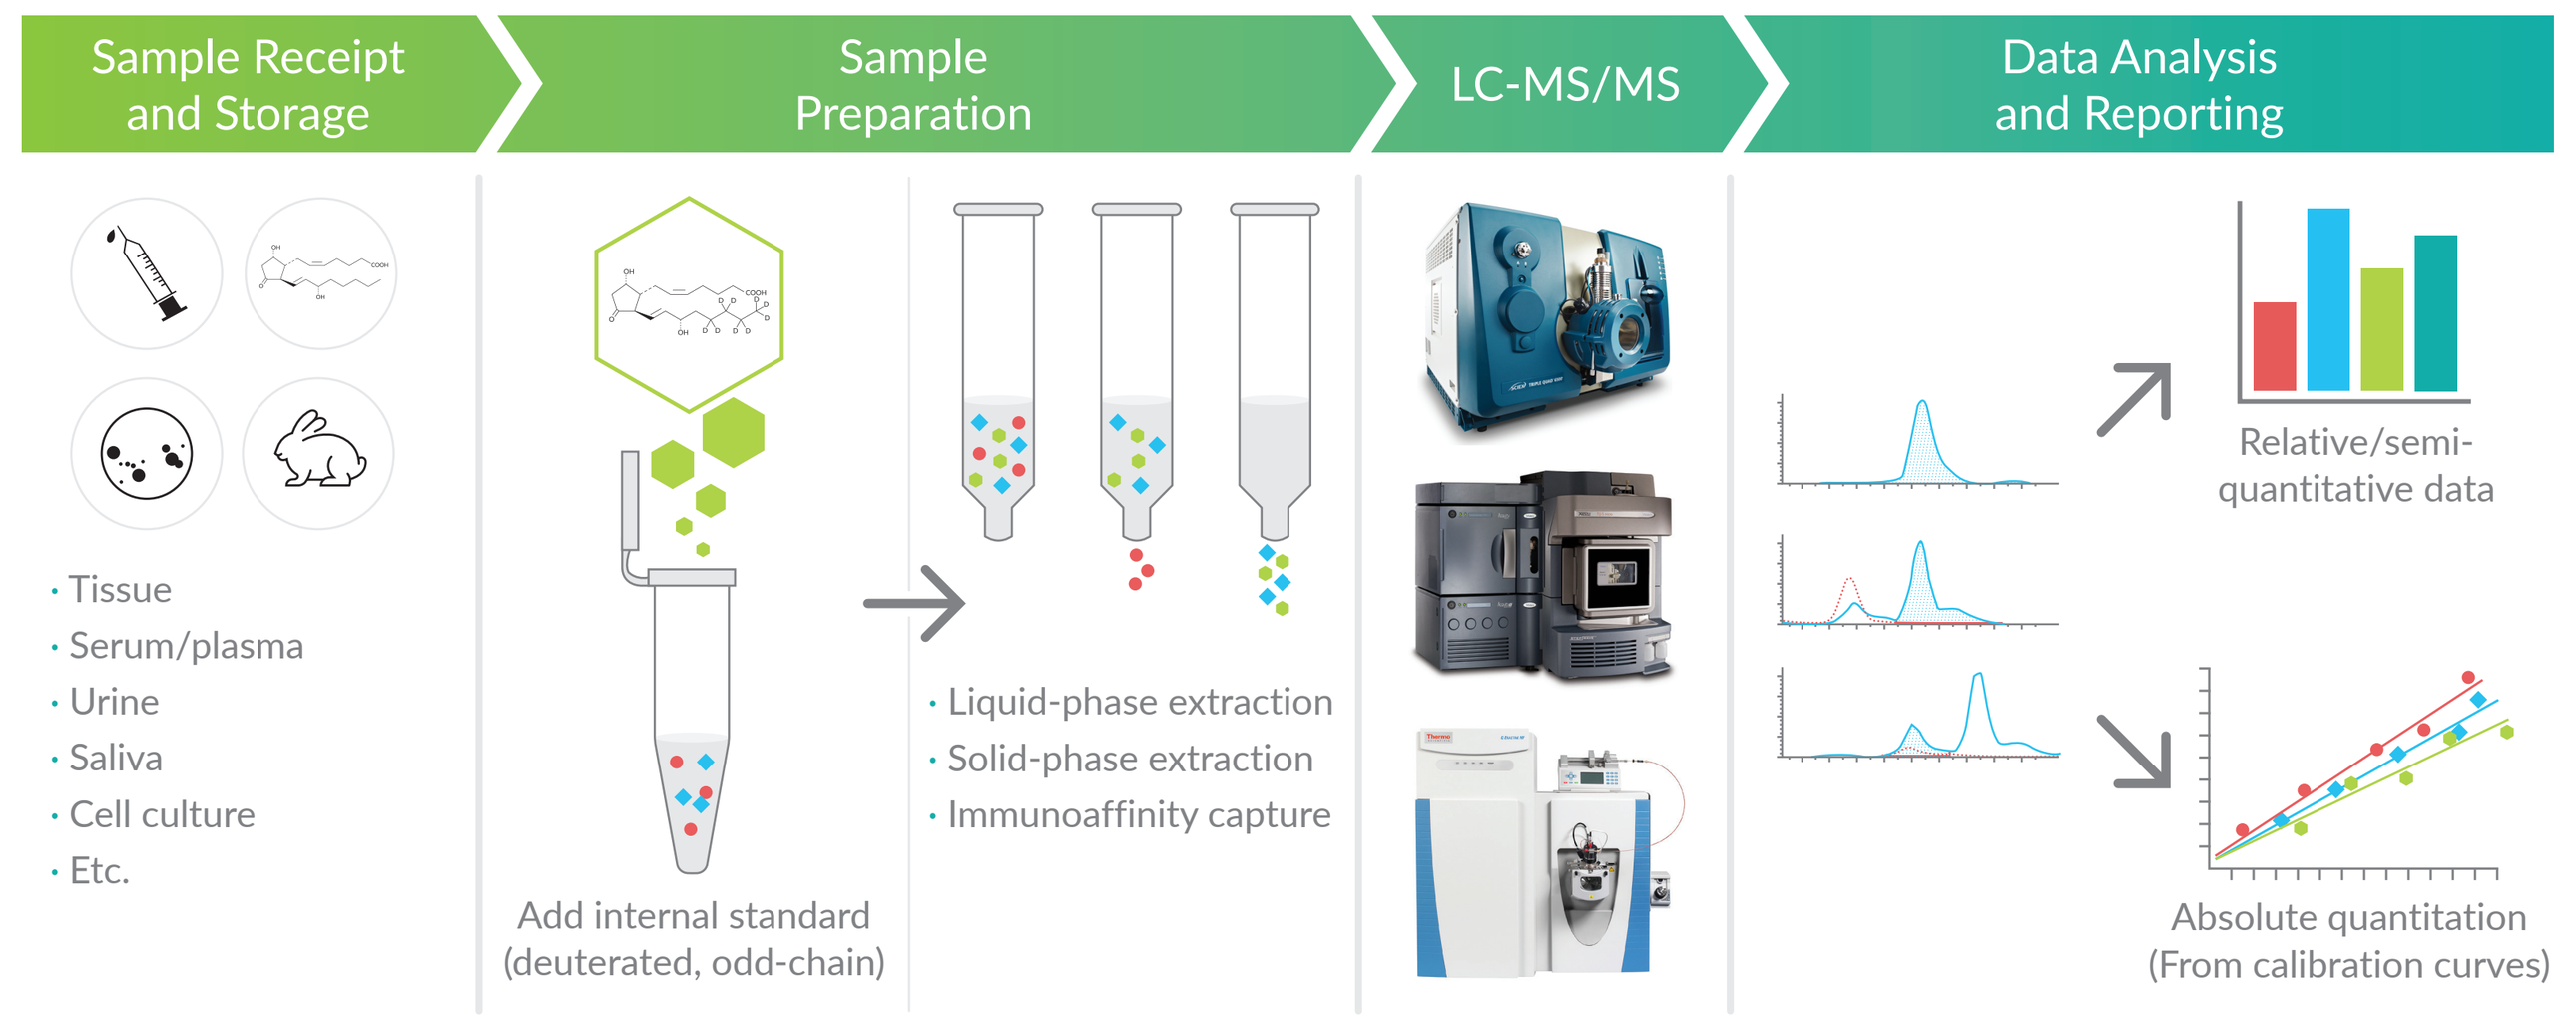

Sample Receipt and Storage
Sample Preparation
LC-MS/MS
Data Analysis and Reporting
· Tissue
· Serum/plasma
· Urine
· Saliva
· Cell culture
· Etc.
Add internal standard (deuterated, odd-chain)
· Liquid-phase extraction
· Solid-phase extraction
· Immunoaffinity capture
Relative/semi-quantitative data
Absolute quantitation (From calibration curves)

Analytes and Internal Standards

Our analysis service can identify and quantify lipids derived from arachidonic acid (AA) through cyclooxygenase (COX), 5-lipoxygenase (5-LO) and other LOs, or cytochrome P450 (CYP450), as well as many other oxylipins derived from other fatty acids such as linoleic, eicosapentaenoic, or docosahexaenoic. This list of analytes may be modified to suit the particular needs of any researcher in any experimental model.

COX	5-LO	Other LOs	CYP450	Others (Not from AA)	Internal Standards
6-keto $PGF_{1\alpha}$	LTB_4	LXA_4	20-HETE	9-HODE	6-keto $PGF_{1\alpha}$-d_4
TXB_2	LTC_4	LXB_4	5(6)-EET	13-HODE	TXB_2-d_4
$PGF_{2\alpha}$	LTD_4	8-HETE	8(9)-EET	17-hydroxy DHA	PGE_2-d_4
PGE_2	LTE_4	9-HETE	11(12)-EET	RvD1	LTD_4-d_5
PGD_2	20-hydroxy LTB_4	11-HETE	14(15)-EET	RvD4	LXA_4-d_5
PGA_2	20-carboxy LTB_4	12-HETE	5(6)-DiHET	RvE1	LTB_4-d_4
PGJ_2	6-*trans* LTB_4	15-HETE	8(9)-DiHET	MaR1	20-HETE-d_6
15-deoxy PGJ_2	5(6)-DiHETE		11(12)-DiHET	PD_1	14(15)-DiHET-d_{11}
12-HHTrE	5-HETE		14(15)-DiHET		9-HODE-d_4
	5-OxoETE				5-HETE-d_8

LC-MS/MS as a Reliable Analytical Tool

Typical LC-MS/MS chromatogram showing the elution profile of oxylipin standards (10 pg) and, where used, deuterated internal standards (100 pg) on a reversed-phase HPLC column. Specific mass-to-charge (*m/z*) transitions are used in the multiple reaction monitoring (MRM) mode to simultaneously detect all analytes.

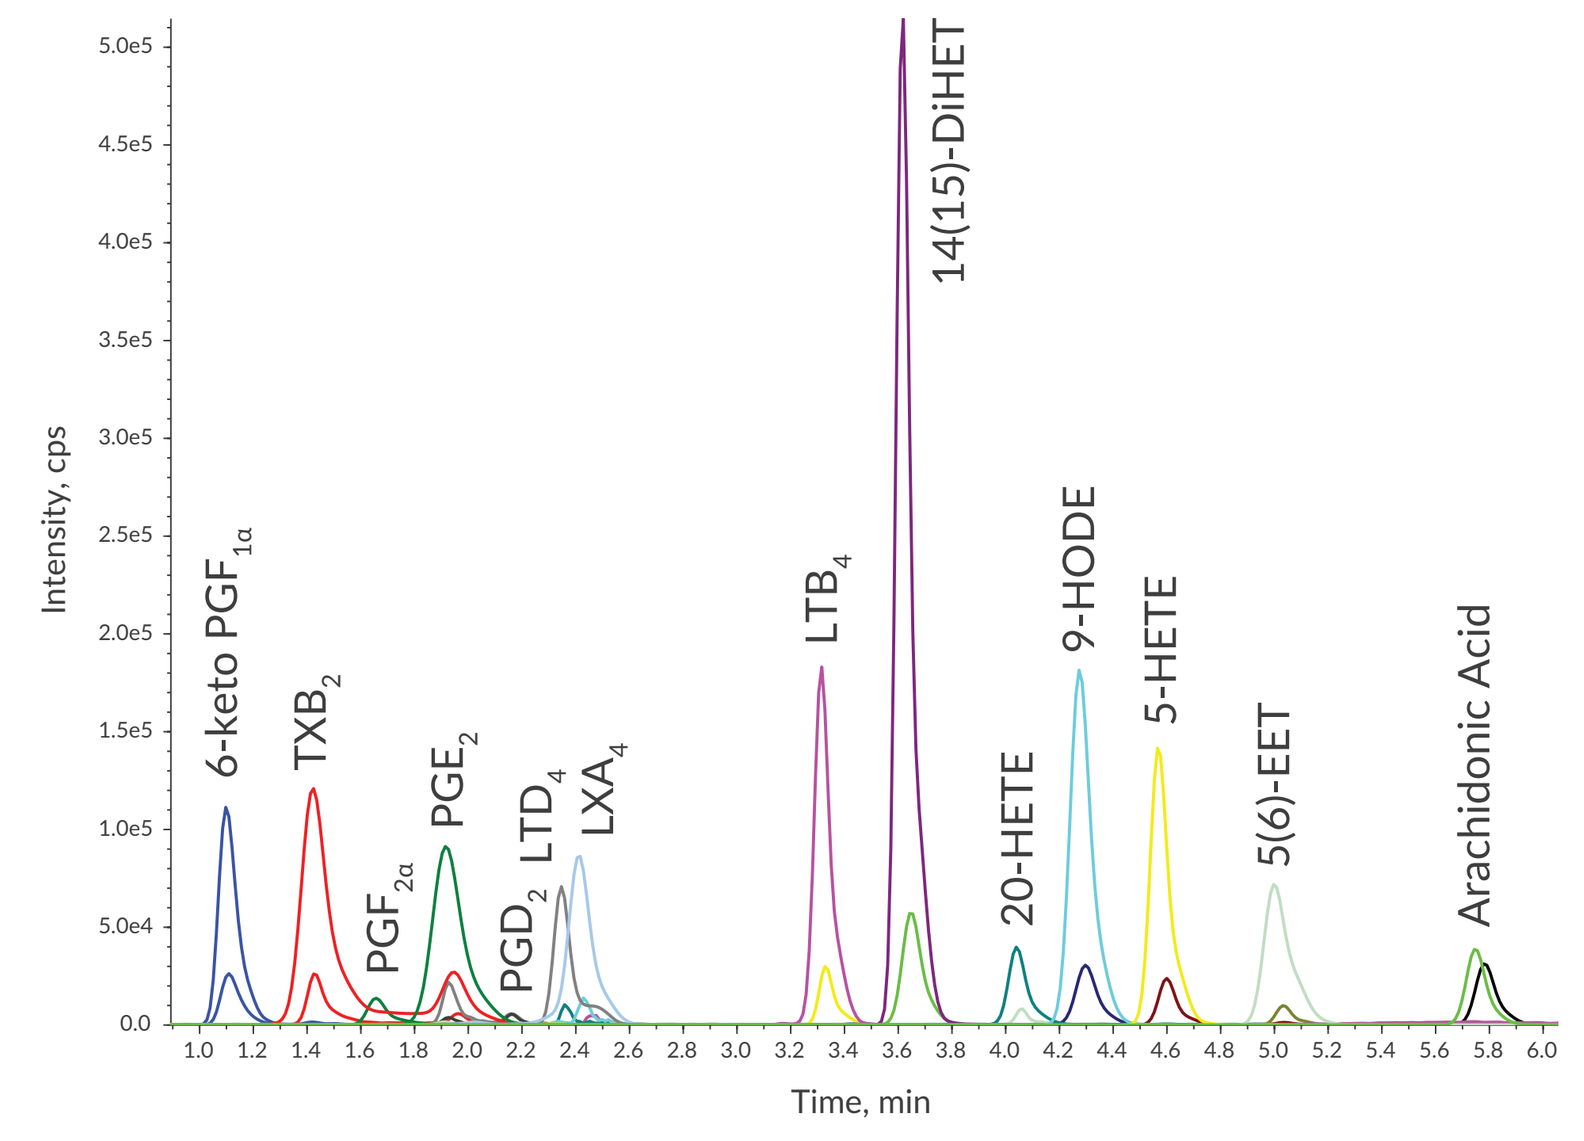

Intensity, cps
5.0e5
4.5e5
4.0e5
3.5e5
3.0e5
2.5e5
2.0e5
1.5e5
1.0e5
5.0e4
0.0
6-keto PGF1α
TXB2
PGF2α
PGE2
PGD2
LTD4
LXA4
LTB4
14(15)-DiHET
20-HETE
9-HODE
5-HETE
5(6)-EET
Arachidonic Acid
1.0 1.2 1.4 1.6 1.8 2.0 2.2 2.4 2.6 2.8 3.0 3.2 3.4 3.6 3.8 4.0 4.2 4.4 4.6 4.8 5.0 5.2 5.4 5.6 5.8 6.0
Time, min

Quantitative Results

Absolute quantitation can be performed using normalized response ratios. Cayman uses high-quality deuterium-labeled internal standards, as well as unlabeled standards, to build calibration curves used in the accurate quantitation of a wide variety of eicosanoids and other oxylipins.

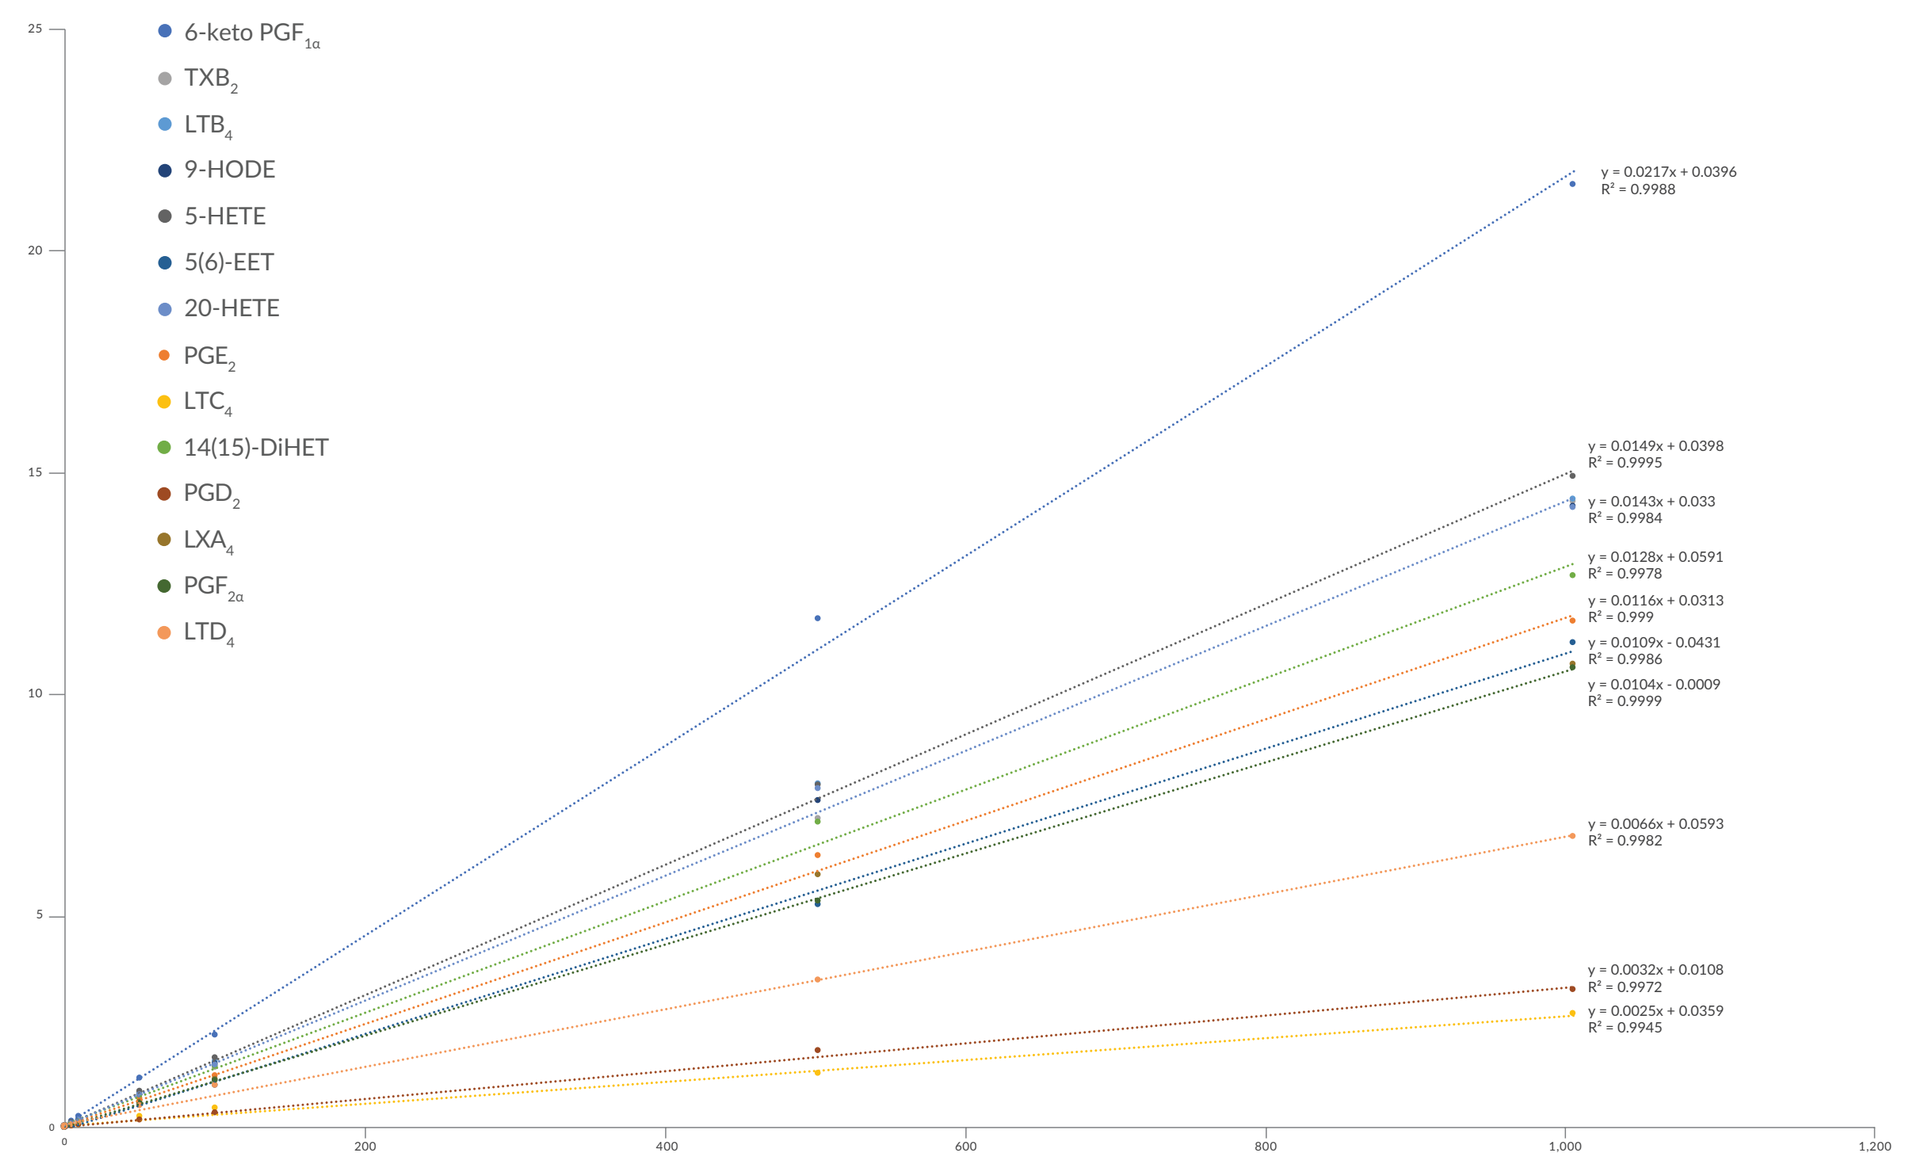

6-keto PGF1α
TXB2
LTB4
9-HODE
5-HETE
5(6)-EET
20-HETE
PGE2
LTC4
14(15)-DiHET
PGD2
LXA4
PGF2α
LTD4
y = 0.0217x + 0.0396
R² = 0.9988
y = 0.0149x + 0.0398
R² = 0.9995
y = 0.0143x + 0.033
R² = 0.9984
y = 0.0128x + 0.0591
R² = 0.9978
y = 0.0116x + 0.0313
R² = 0.999
y = 0.0109x - 0.0431
R² = 0.9986
y = 0.0104x - 0.0009
R² = 0.9999
y = 0.0066x + 0.0593
R² = 0.9982
y = 0.0032x + 0.0108
R² = 0.9972
y = 0.0025x + 0.0359
R² = 0.9945
200
400
600
800
1,000
1,200

Analytical Accuracy and Reproducibility

Cayman is committed to the use of high-quality analytical standards and confidently presents time-tested methodology to ensure quantitative accuracy. Interlaboratory evaluation of quantitative results for serum LTB_4 between Cayman and a collaborating lipidomics laboratory performing a similar LC-MS/MS method on duplicate serum samples demonstrates excellent correlation.

Interlaboratory Correlation for Serum LTB_4

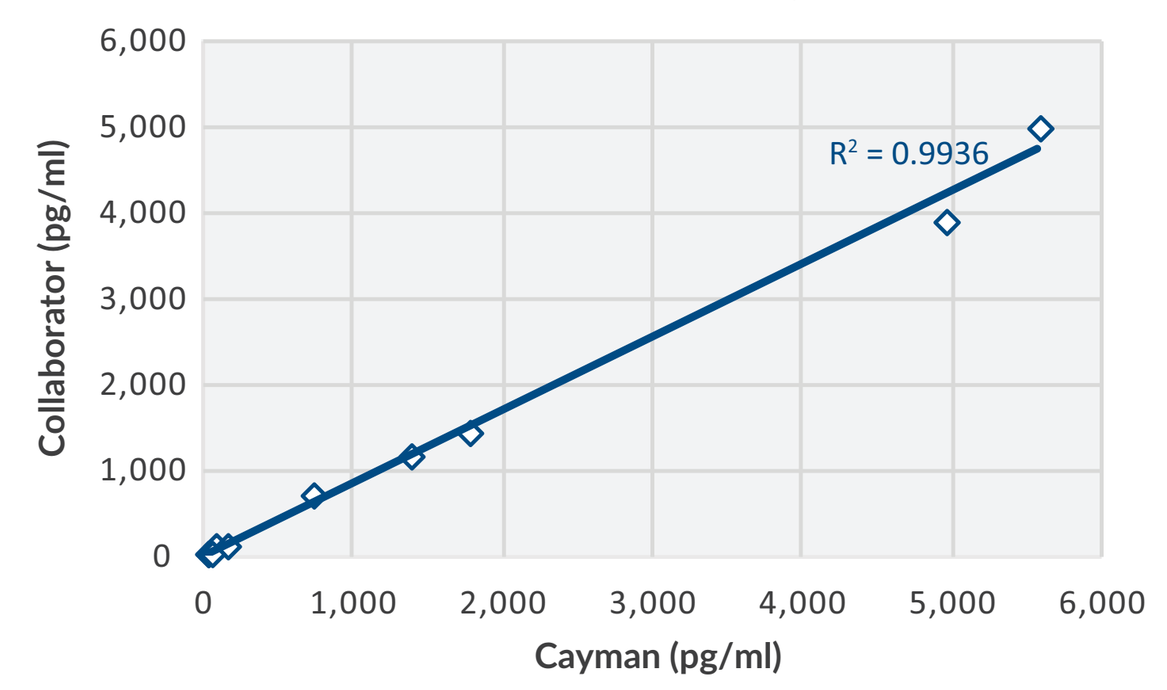

Collaborator (pg/ml)
6,000
5,000
4,000
3,000
2,000
1,000
0
R² = 0.9936
0 1,000 2,000 3,000 4,000 5,000 6,000
Cayman (pg/ml)

Results You Can Trust

- **Expert consultation on best practices for sample collection, pre-shipment processing, and storage to preserve sample integrity**
- **Optimized sample extraction protocols guided by accepted literature methods (liquid-liquid extraction, solid-phase extraction, ztargeted immunoaffinity capture)**
- **Semi-quantitative profile analysis, with option to follow up with absolute quantitation of customizable analyte sets**
- **Timely updates, interactive real-time data review, and comprehensive reporting with strict confidentiality**

Cell Culture and Biological Fluids

Cayman's scientists can support your cell culture studies from assay design and development to analysis, and have the experience to measure lipids in common and more complex matrices. The below example shows MRM chromatograms (blue traces) of LTB_4, LTC_4, LTD_4, and LTE_4 in vehicle control (top row) and replicate ionophore-stimulated (middle and bottom rows) mouse bone marrow-derived mast cells. Red traces correspond to the deuterium-labeled internal standards, LTD_4-d_5 (three left columns) or LTB_4-d_4 (right column).

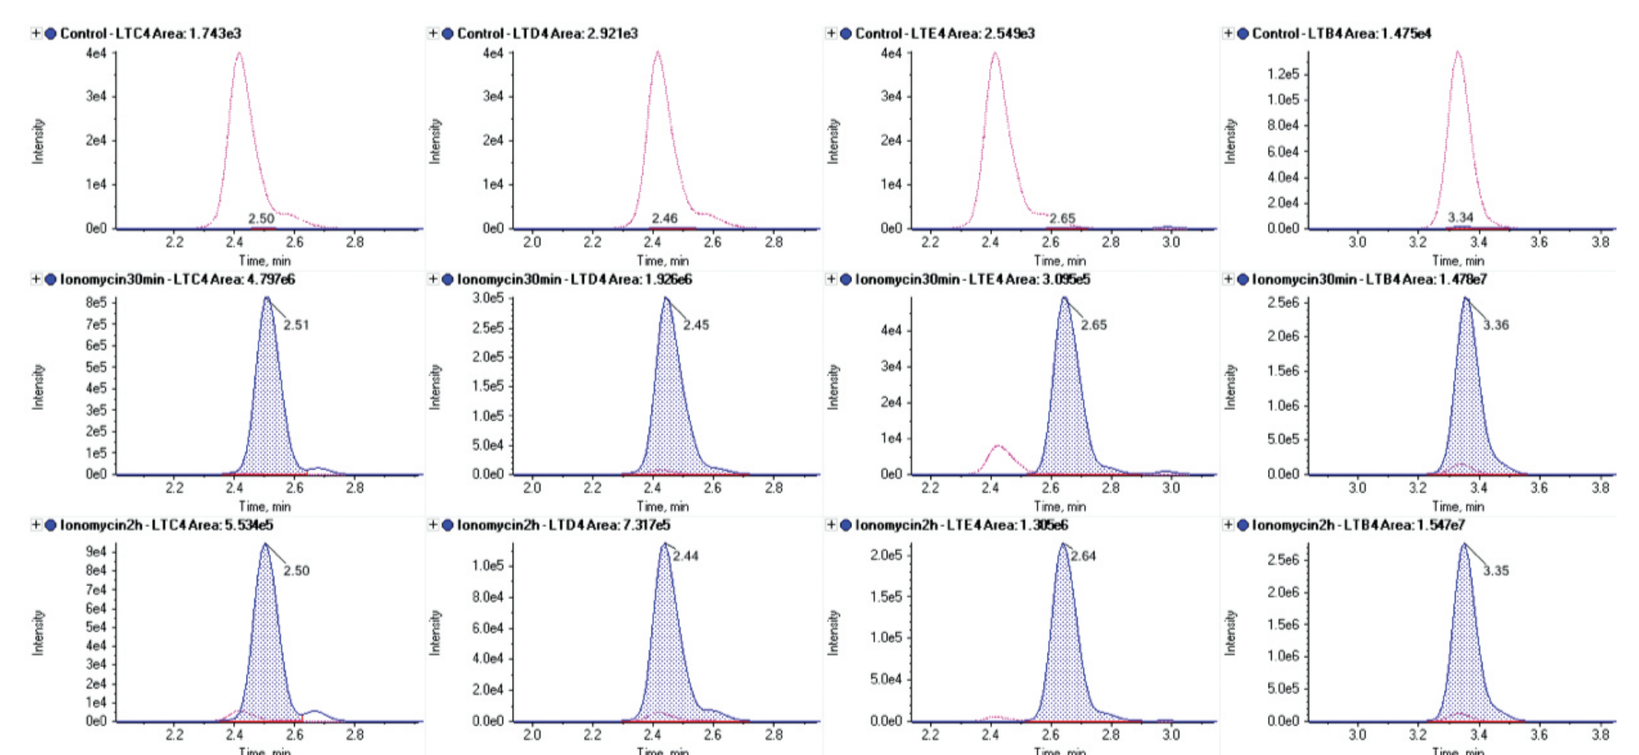

Biological fluids such as plasma, serum, or urine are among the most accessible samples available for human and animal research. This table shows MRM *m/z* transitions, chromatographic and integration information, and calculated concentrations of selected lipids from an extract of 20 µl of commercial mouse plasma. It demonstrates the ability to quantify a variety of bioactive lipids over a wide dynamic range (five orders of magnitude in this case) in the same sample.

Analyte	MRM Info	Area	Area/IS Ratio	Retention Time	Signal/Noise	Calculated Concentration (pg/ml)
TXB_2	369.2 / 169.1	2.62E+05	0.33	1.51	353.5	23.0
PGD_2	351.2 / 271.2	4.83E+05	0.6	2.07	93.1	50.7
LTB_4	335.2 / 195.1	5.30E+03	0.01	3.33	17.8	0.33
5-HETE	319.2 / 115.1	1.30E+05	0.39	4.62	435.4	26.0
14(15)-DiHET	337.2 / 207.1	3.73E+05	1.13	3.66	1,011.7	86.0
9-HODE	295.2 / 171.1	1.33E+06	4.02	4.3	1,196.4	279
AA	303.2 / 205.1	3.25E+07	98.42	5.79	14,112	7,155

Learn more about our service capabilities at

www.caymanchem.com/services

Animal Models

Tissue samples from a range of animal models can be processed and analyzed. The figures below show comparative data between spleens from healthy mice *versus* spleens from mice with experimental autoimmune encephalitis (EAE). Using standard statistical analysis tools, complex mass spectrometry data for dozens of bioactive lipids could be analyzed to identify a changing trend in the 5-lipoxygenase pathway products.

Complex Raw Data

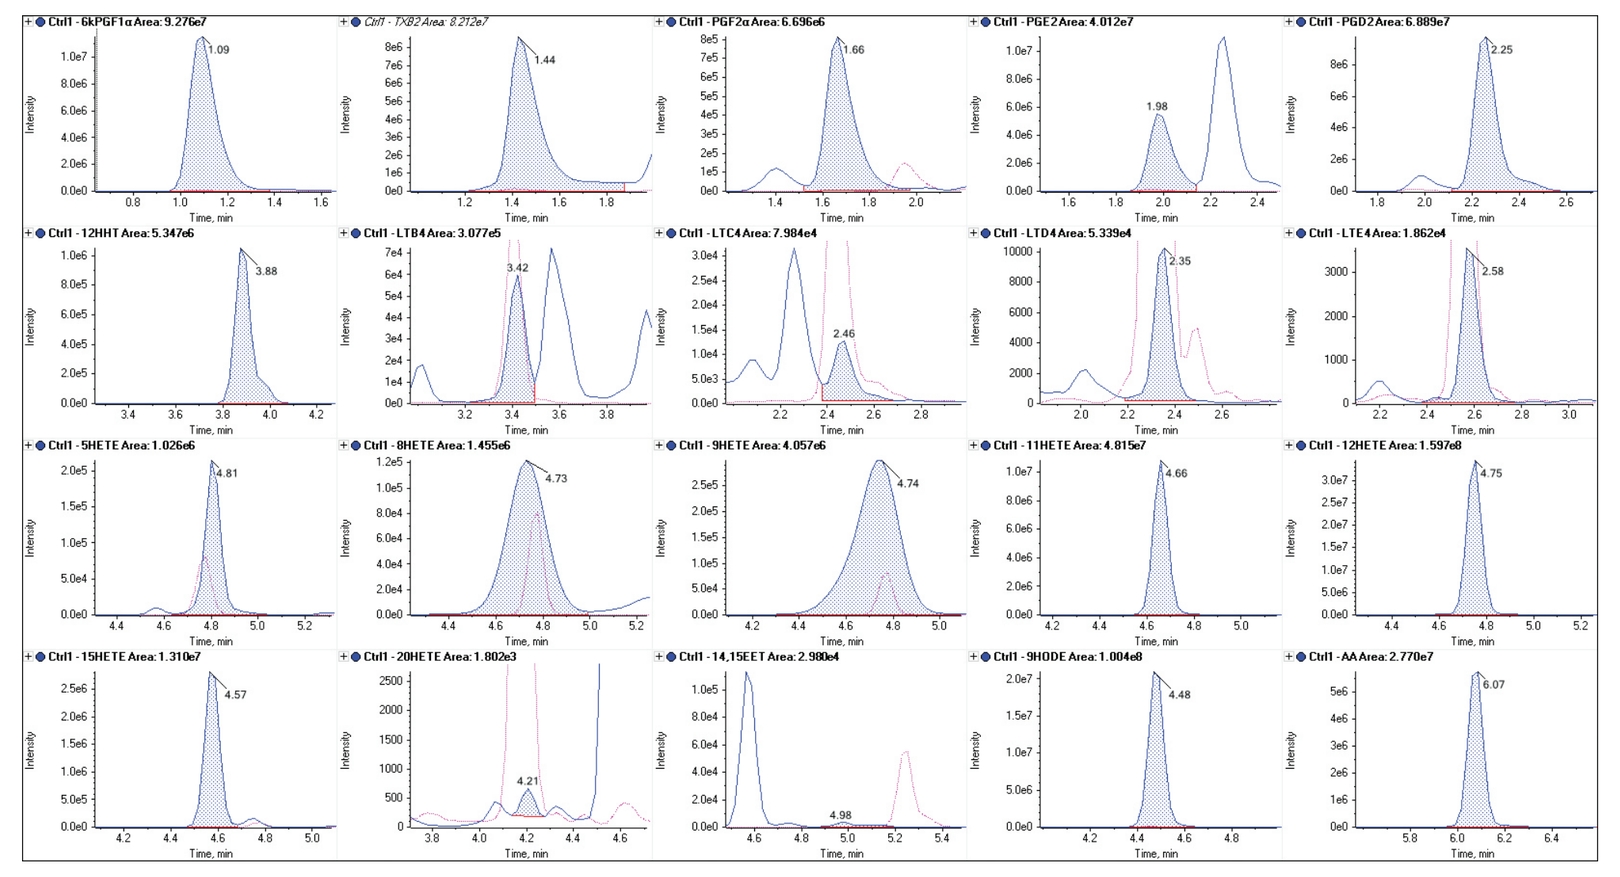

Large sets of chromatograms are quickly reviewed for accuracy and compound identity is verified. This data may be reviewed and shared with customers in real time upon request.

Area Ratios Analyte/IS	Healthy 1	Healthy 2	Healthy 3	EAE 1	EAE 2	EAE 3
6-keto $PGF_{1\alpha}$	148.80	130.39	284.76	103.64	59.89	103.17
TXB_2	66.91	55.64	118.08	60.12	48.53	53.21
$PGF_{2\alpha}$	6.42	5.80	14.05	7.49	9.43	9.89
PGE_2	37.15	28.28	90.95	21.93	24.44	20.27
PGD_2	50.75	46.54	102.62	78.77	72.94	71.85
12-HHTrE	13.72	19.33	34.83	15.25	12.26	14.76
LTB_4	0.75	0.82	1.86	6.95	6.30	4.39
LTC_4	0.18	0.06	0.48	1.67	1.43	1.26
LTD_4	0.07	0.03	0.21	1.14	0.67	0.64
LTE_4	0.27	0.17	1.32	2.54	1.96	1.68
5-HETE	2.68	2.38	5.73	23.25	18.99	15.20
8-HETE	3.33	7.13	6.24	3.27	3.56	4.06
9-HETE	9.80	21.03	20.62	11.80	12.63	14.43
11-HETE	117.38	121.87	236.75	70.89	80.65	92.64
12-HETE	382.33	900.38	774.85	454.97	511.08	603.47
15-HETE	30.59	25.88	61.73	32.46	28.47	29.89
20-HETE	0.03	0.13	0.03	0.05	0.03	0.03
14(15)-DiHET	0.08	0.10	0.08	0.14	0.15	0.14
9-HODE	145.84	294.31	132.09	79.86	81.66	72.47
AA	376.91	528.27	523.44	852.11	453.98	633.48

Quantitative data may then be calculated from calibration curves. This data is included in final reports.

Data Analysis

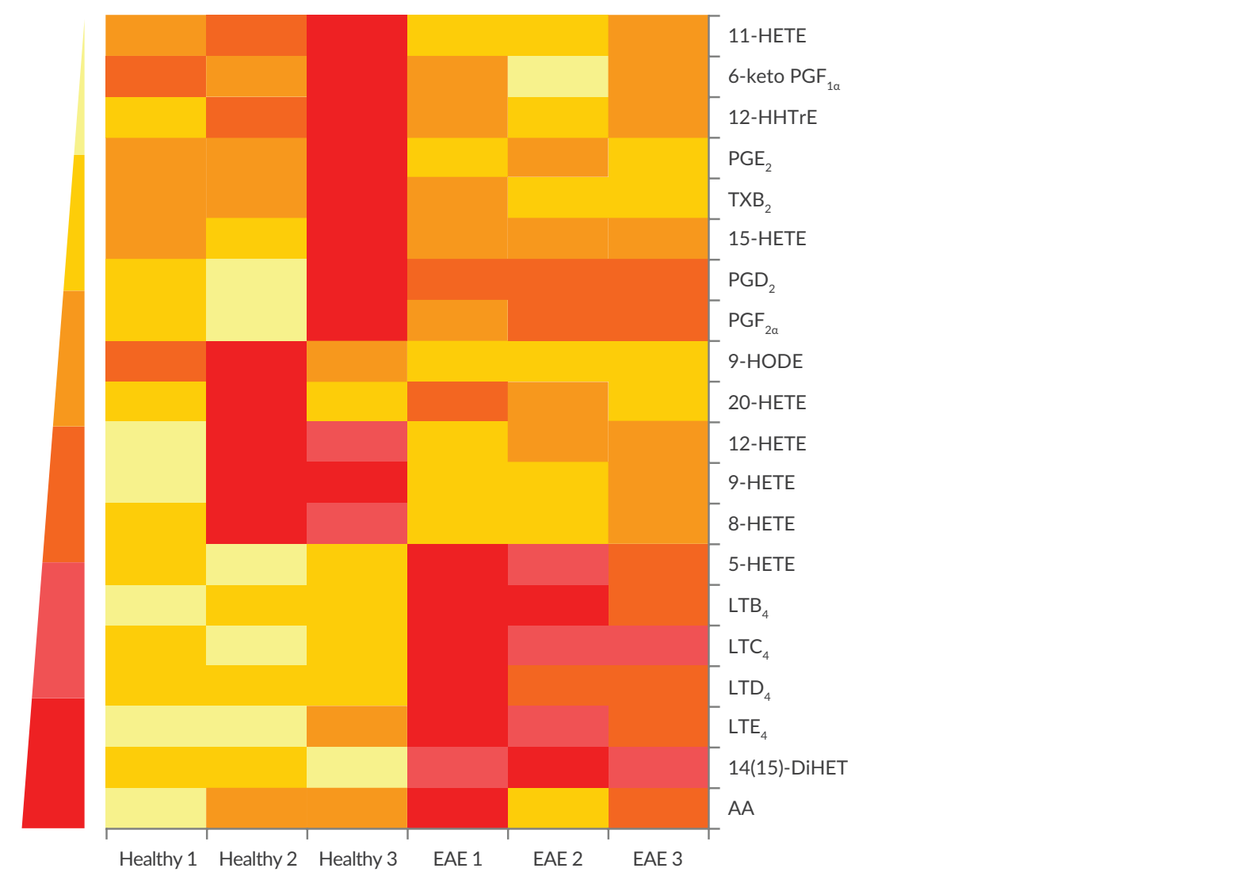

11-HETE
6-keto PGF1α
12-HHTrE
PGE2
TXB2
15-HETE
PGD2
PGF2α
9-HODE
20-HETE
12-HETE
9-HETE
8-HETE
5-HETE
LTB4
LTC4
LTD4
LTE4
14(15)-DiHET
AA
Healthy 1
Healthy 2
Healthy 3
EAE 1
EAE 2
EAE 3

A heat map plot or other statistical visualization tools (volcano plots, PCA plots) can be generated to quickly cluster important changes, allowing rapid pinpointing of important trends.

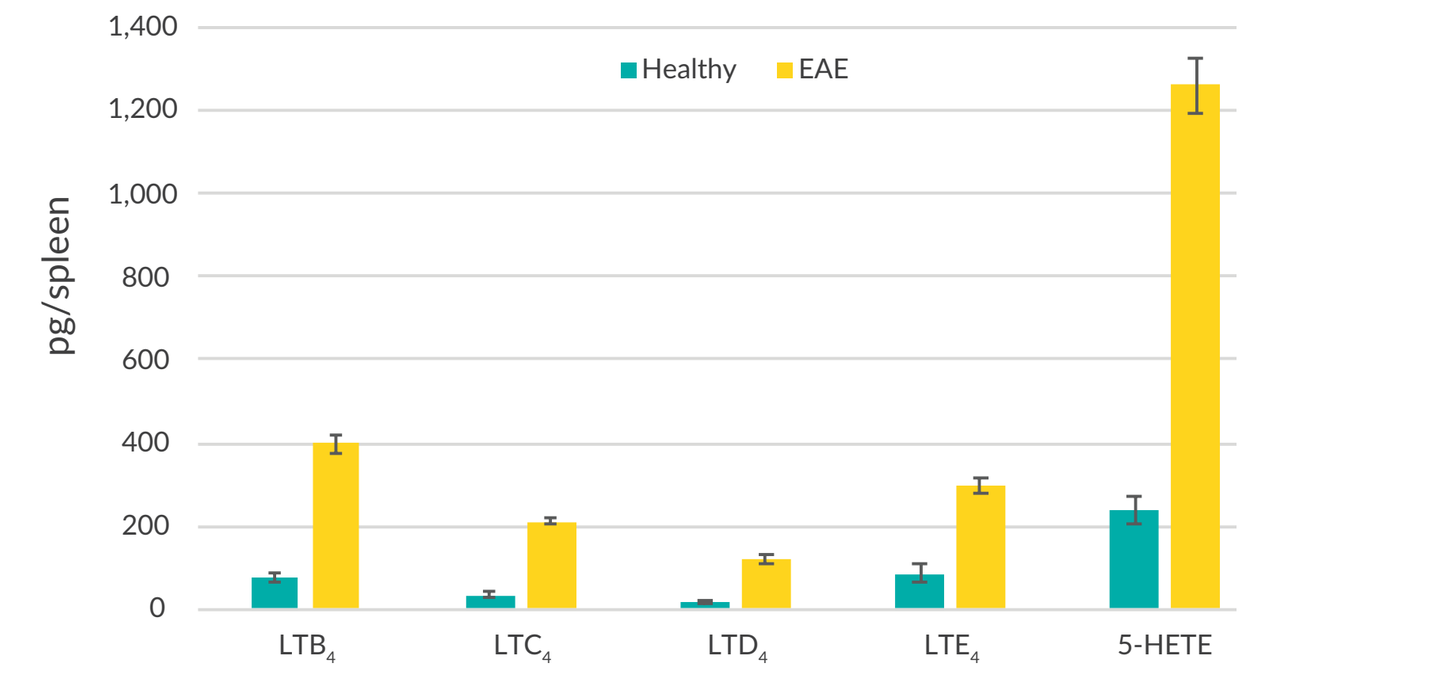

pg/spleen
1,400
1,200
1,000
800
600
400
200
0
Healthy
EAE
LTB4
LTC4
LTD4
LTE4
5-HETE

Focused data analysis and review of trends allows for adequate interpretation of the data and generation of new hypotheses.

Conclusions and New Hypotheses

Acknowledgment: We would like to thank Dr. Athena Soulika (Associate Professor, UC Davis School of Medicine and Investigator, Institute of Pediatric Regenerative Medicine, Shriners Hospitals for Children) for providing cell and tissue samples.